Computational Systems Pharmacology of Antibody-Drug Conjugates

Inez Lam[1], K. Phin Chooi[2], Conor S. Barry[2], Balakumar Vijayakrishnan[2], Thaïs Cailleau[2], Peter C. Tyrer[2], Rosalin Arends[3], Kathryn Ball[3], Feilim Mac Gabhann[1]

[1] Institute for Computational Medicine and Department of Biomedical Engineering, Johns Hopkins University
[2] Spirogen, AstraZeneca
[3] Clinical Pharmacology and Safety Sciences, AstraZeneca

Johns Hopkins University

AstraZeneca

Background

Antibody-Drug Conjugates (ADCs) are engineered immunoconjugate drugs composed of 3 core components: (1) a monoclonal antibody (Ab) and (2) one or more cytotoxic small molecules (known as warheads), attached via (3) a chemical linker. The antibody targets a specific tumor antigen, so the warhead can selectively kill tumor cells while sparing healthy tissue. As a designed, targeted drug delivery vehicle to selected cells, one might expect ADCs to have high success rates; however, so far 55 ADCs have failed and only 5 have been approved for use in oncology, and those that are approved typically only succeed in a fraction of patients[1].

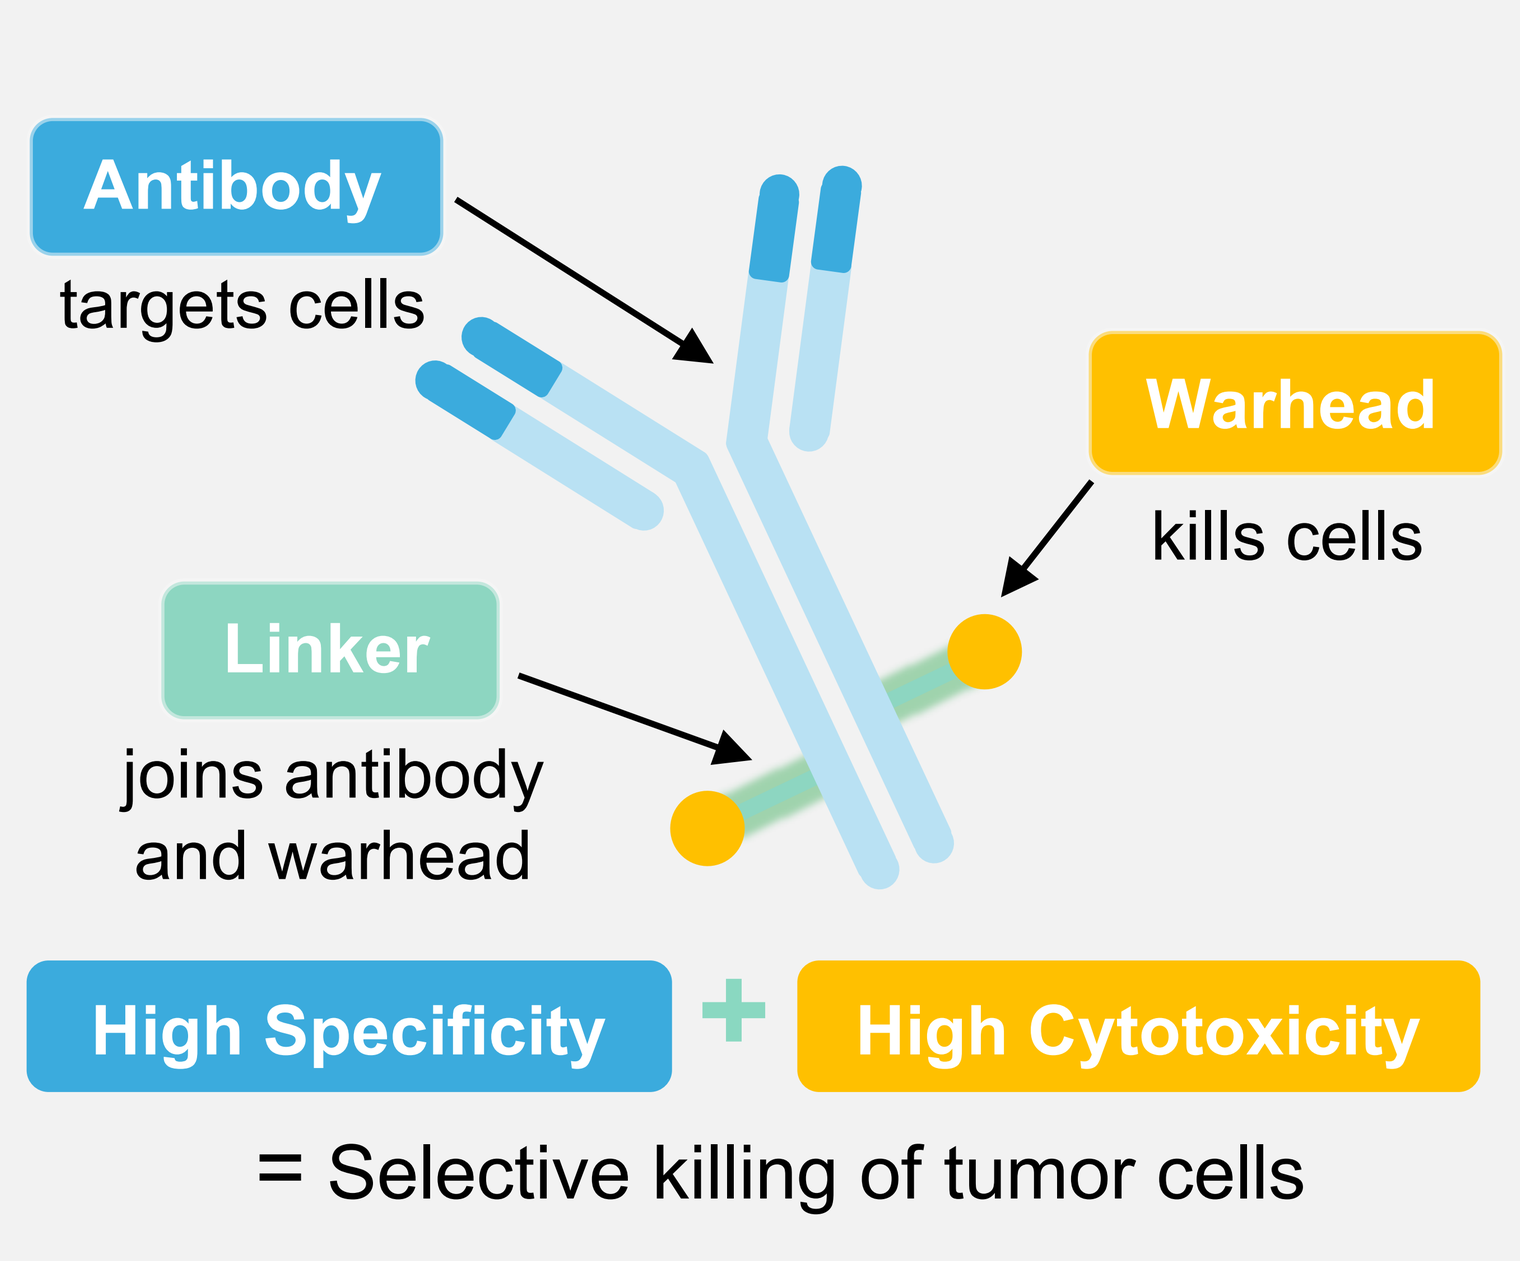
Antibody
targets cells
Warhead
kills cells
Linker
joins antibody and warhead
High Specificity
High Cytotoxicity
= Selective killing of tumor cells

Antibody-Drug Conjugate Structure

With their modular structure, ADCs have multiple design levers with which to optimize efficacy and minimize toxicity. This task calls for a deep understanding of the biological and pharmacological systems, processes, and mechanisms at play. Seeking answers through experimental methods alone can be laborious, expensive, or even impossible. Computational modeling can probe questions and enhance insight through quantitative simulation of drug action and performance. Quantitative systems pharmacology (QSP) approaches integrate mechanistic knowledge with biomedical data at multiple scales to construct an interpretable and predictive model. QSP models use existing biological and clinical data to help us explore mechanisms and hypotheses efficiently, and support decision-making in drug development and clinical practice. Strengthening our understanding of antibody-drug conjugates will support the continued development of improved, innovative targeted cancer treatments.

Methodology

We have constructed a computational systems pharmacology model with cellular and intracellular mechanisms specific to ADCs with pyrrolobenzodiazepine (PBD) warheads to understand the impact of ADC characteristics on in vitro efficacy and toxicity. The model is comprised of coupled, deterministic, nonlinear, ordinary differential equations (ODEs), outputting the concentrations of each molecule or molecular complex over time. Model parameters were estimated from literature, or calibrated and validated using in vitro experimental data on cytotoxicity provided by AstraZeneca for anti-HER2-PBD ADCs; the antibodies target HER2, but carry payloads (the attached linkers and warheads) with differing properties. The model describes ADC binding, internalization, recycling, intracellular trafficking and degradation, warhead binding, efflux and influx of warhead.

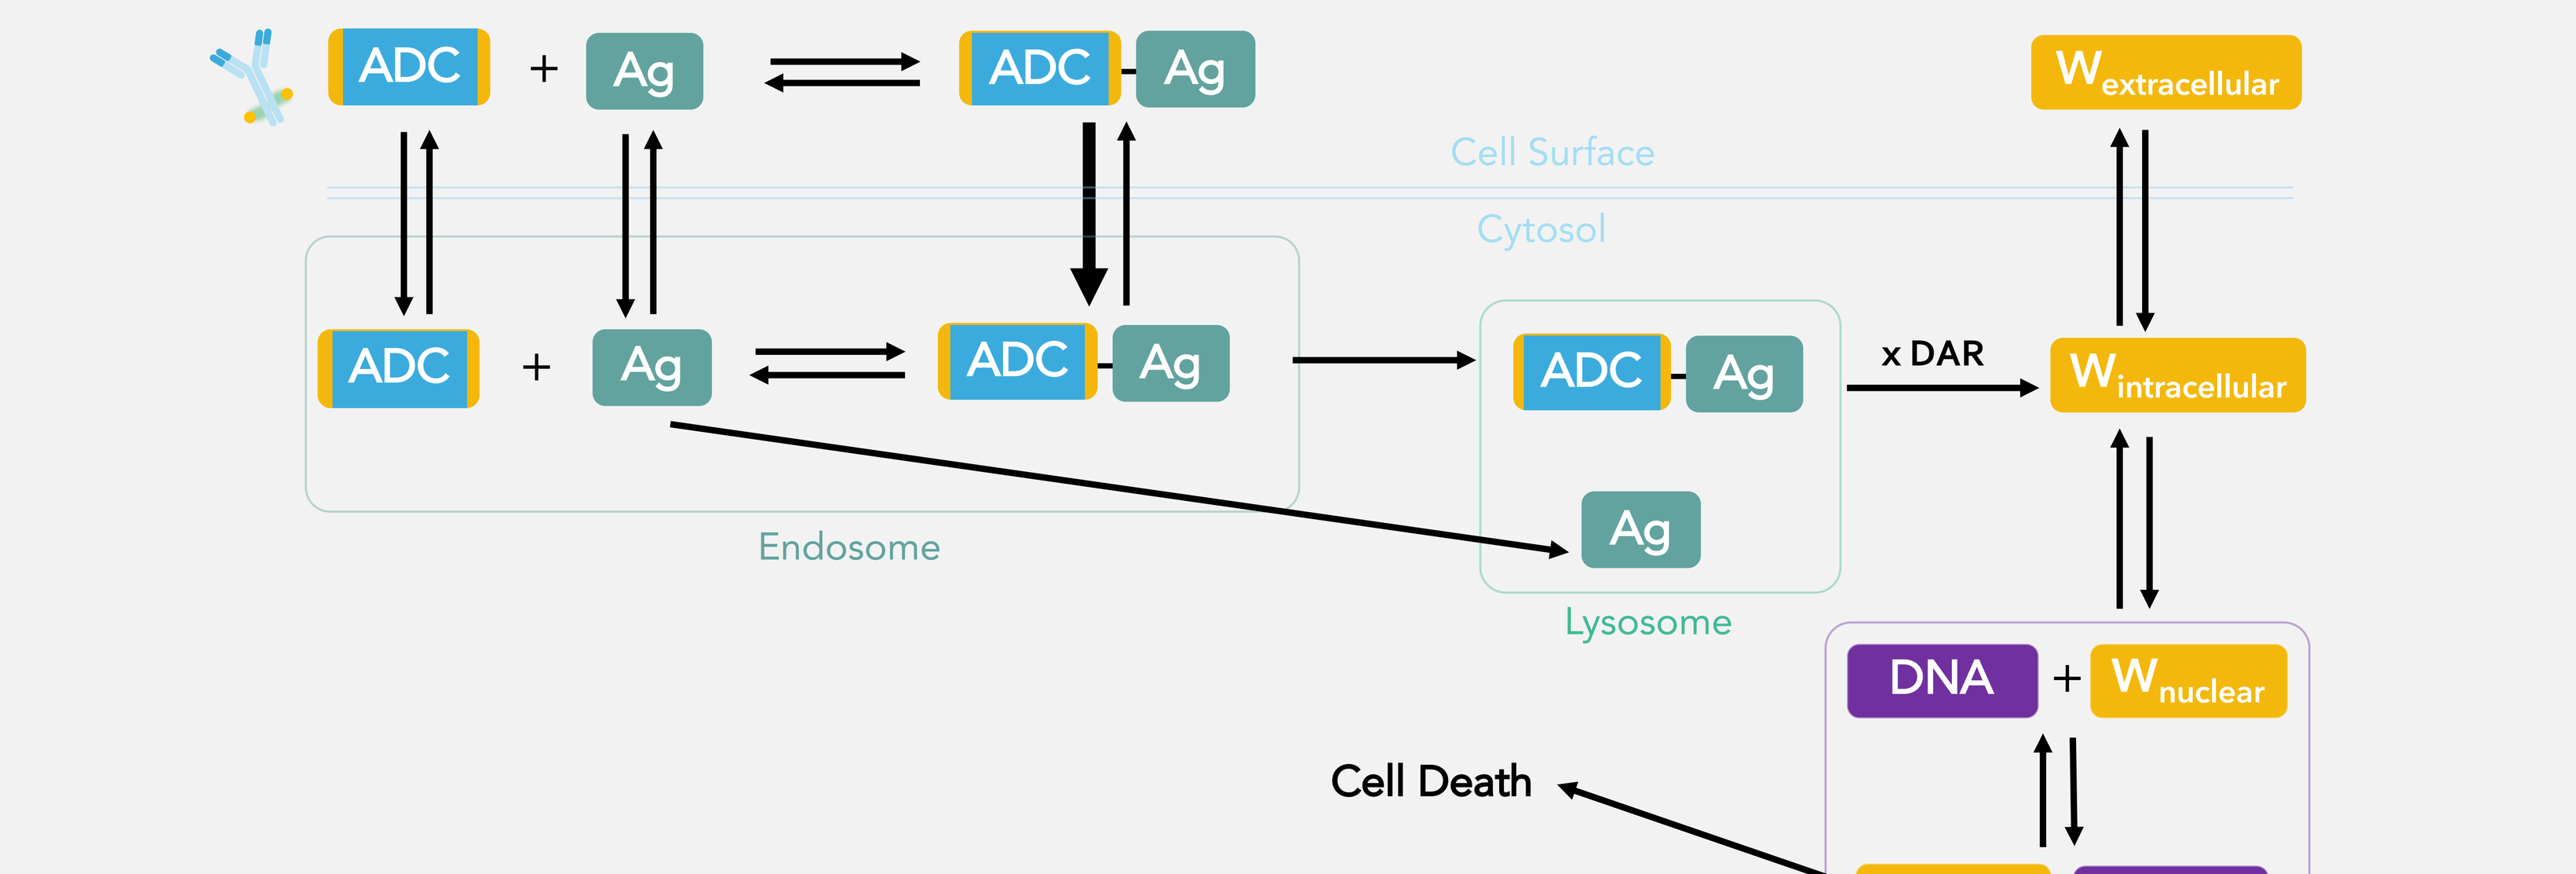
ADC
Ag
Cell Surface
Cytosol
Endosome
Lysosome
x DAR
W extracellular
W intracellular
DNA
W nuclear
Cell Death
Nucleus

In Vitro Model Schematic

Exploring ADC Design Properties

Drug-to-Antibody Ratio

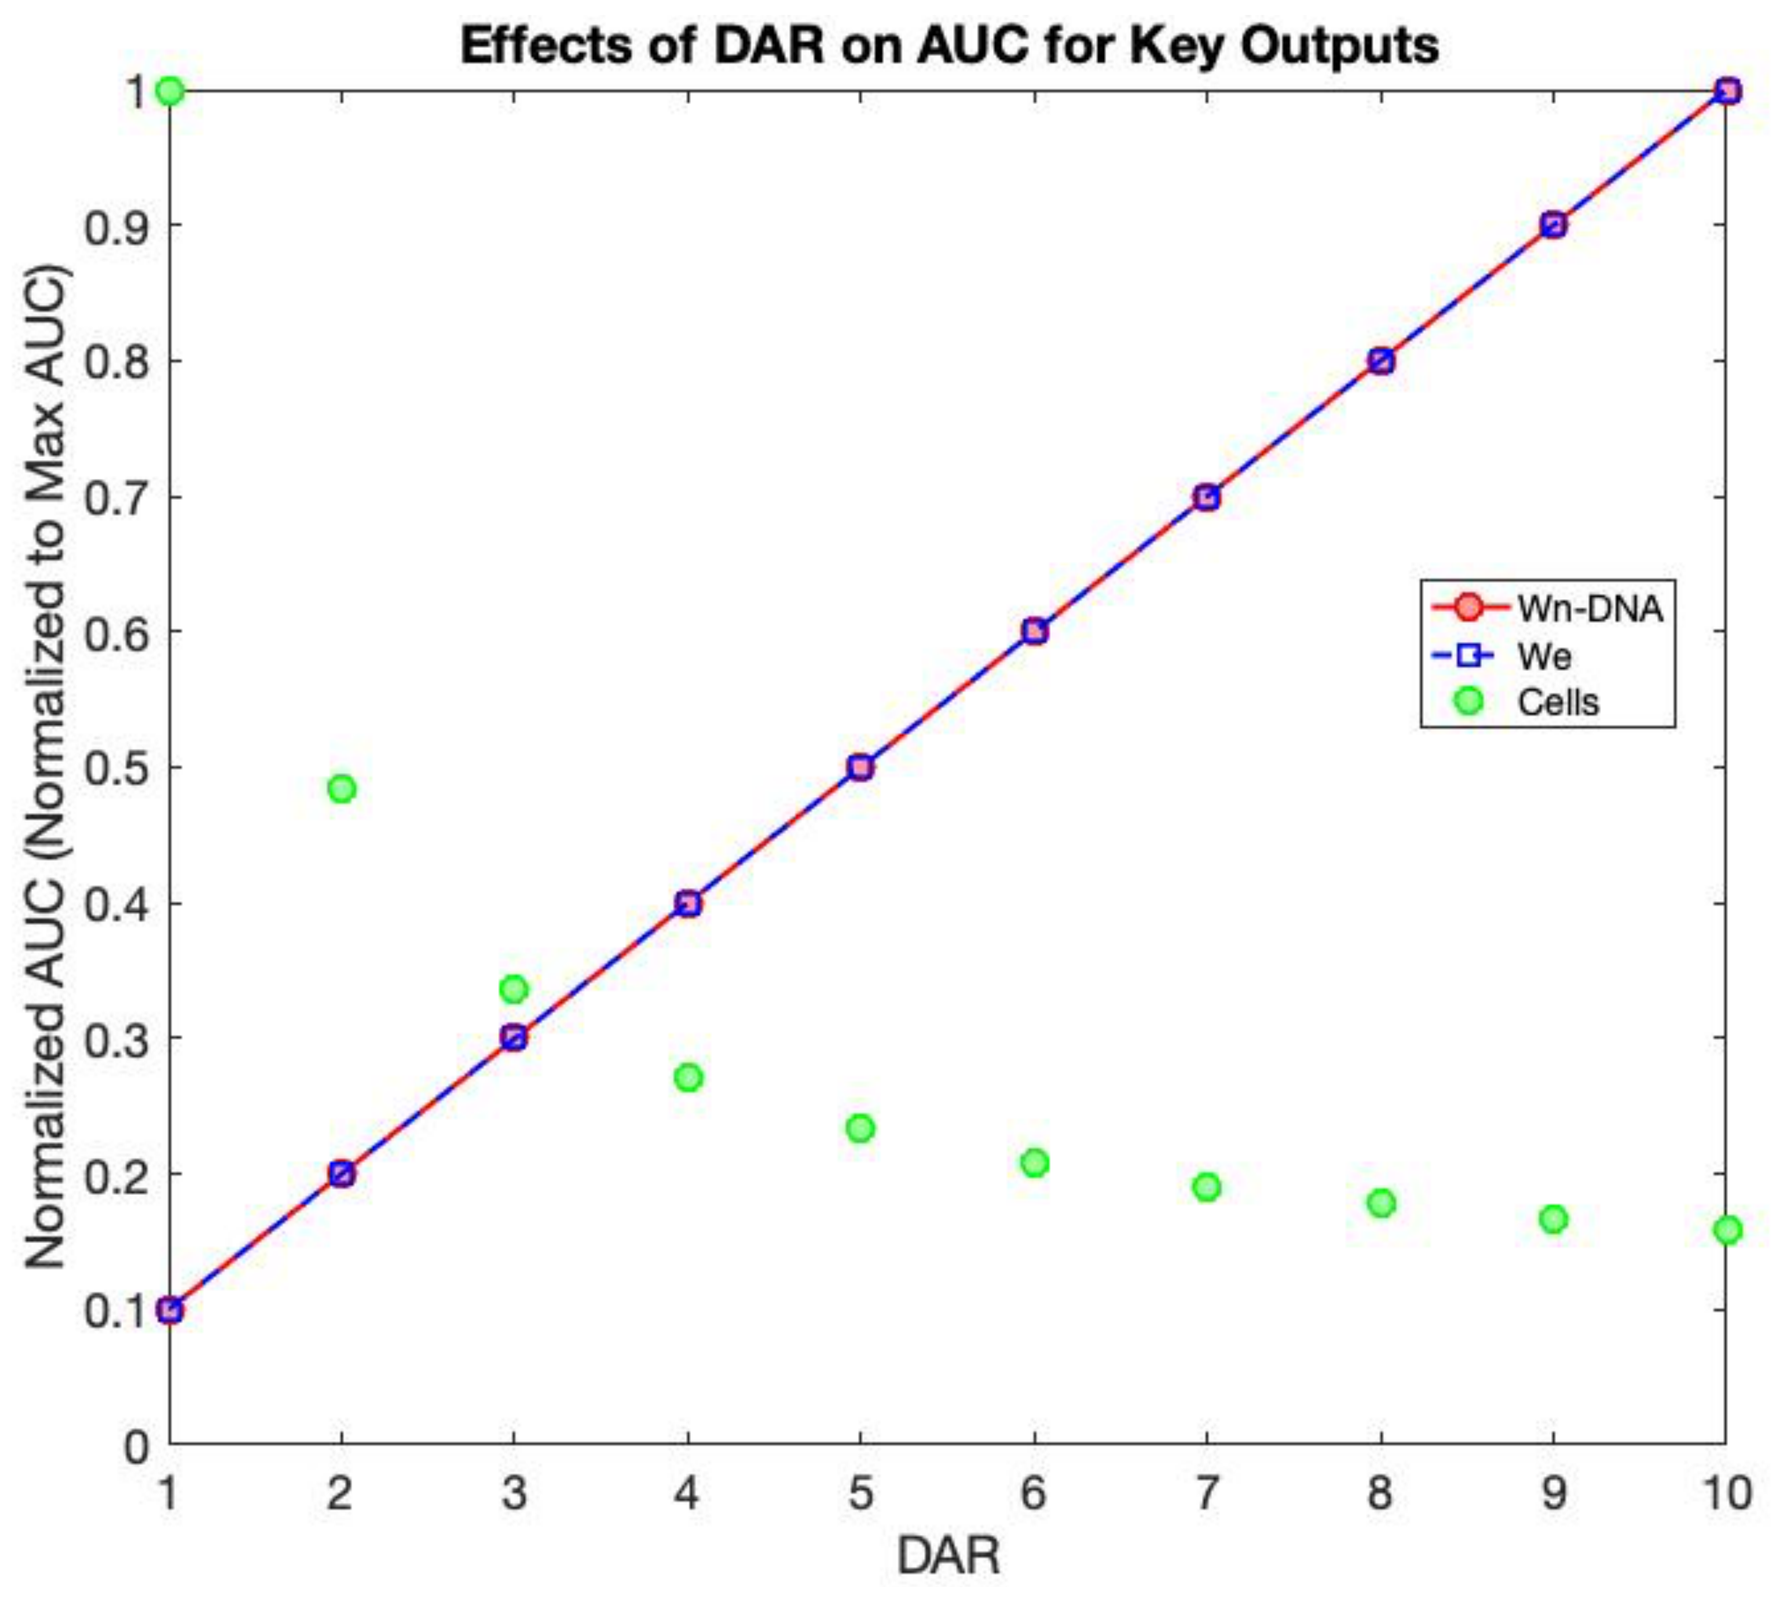
Effects of DAR on AUC for Key Outputs
Normalized AUC (Normalized to Max AUC)
DAR
Wn-DNA
We
Cells

Changes to the Drug-to-Antibody Ratio (DAR) cause linear increases in warhead levels and exponential decreases in cell population. The number of warheads per antibody is a key property in ADC design. As DAR increases, the areas under the curve (AUC) for warhead-DNA complex and extracellular warhead increase linearly, while the AUC for cell population decreases exponentially. The biggest gain in cell killing results from first few warhead molecules, suggesting that the optimal DAR for this system may be between 2 and 4 to maximize efficacy while minimizing toxicity.

Warhead Potency

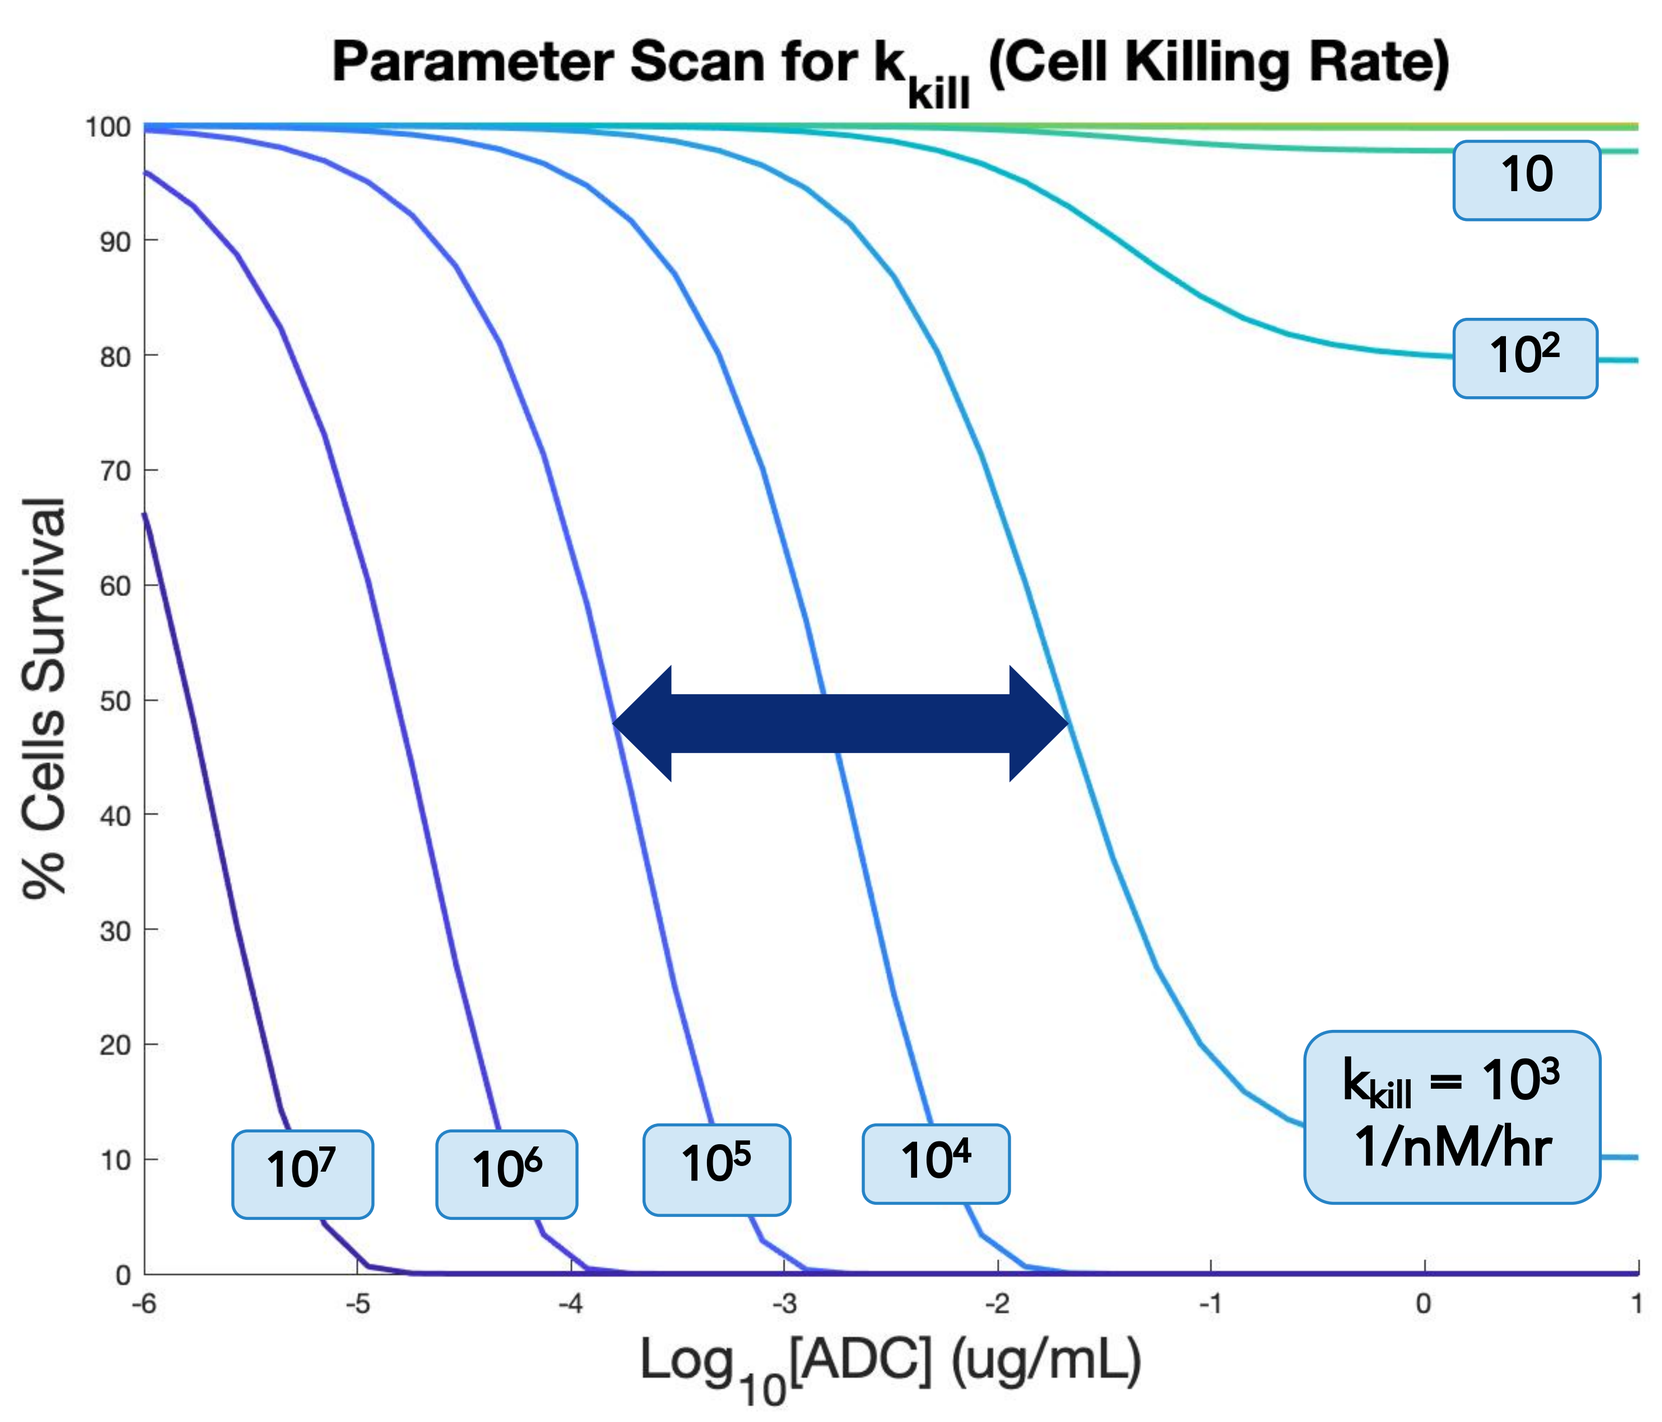
Parameter Scan for k kill (Cell Killing Rate)
% Cells Survival
Log10[ADC] (ug/mL)
10
10²
k kill = 10³ 1/nM/hr
10⁷
10⁶
10⁵
10⁴

Parameter scan reveals most likely range for warhead potency. We conducted a parameter scan for the cell killing rate (k_{kill}) by simulating with parameter values at different orders of magnitude (between 10^{-2} – 10^{7}) to see impact on cell survival percentage. The cell killing rates for PBD-ADCs fell within the range indicated by the blue arrow (k_{kill} between 10^3 – 10^5), suggesting that these rates can be used as a reflection of warhead potency (a key ADC design property) for this system of ADCs.

Sensitivity Analysis

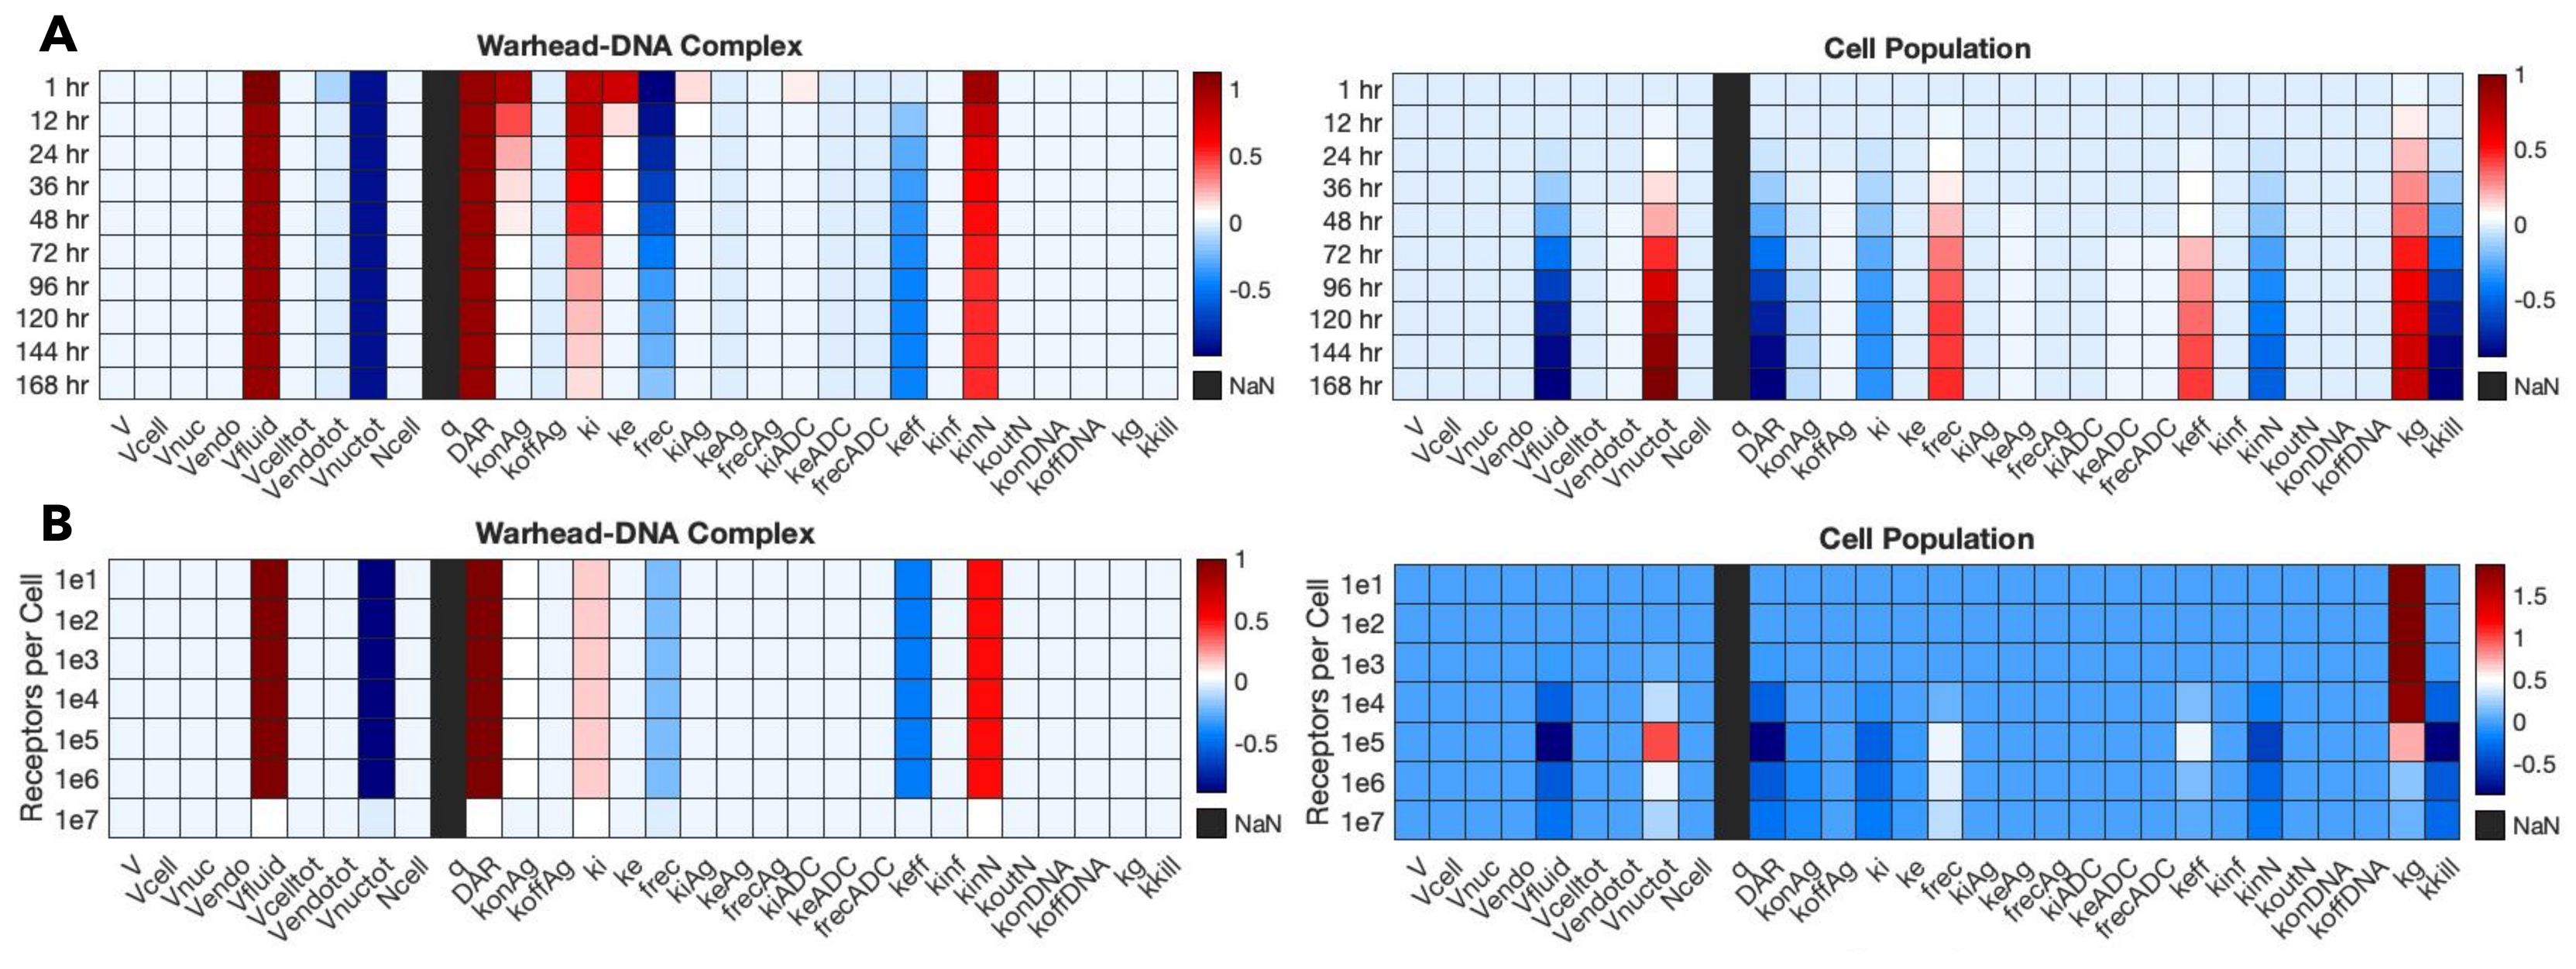
A
Warhead-DNA Complex
Cell Population
1 hr
12 hr
24 hr
36 hr
48 hr
72 hr
96 hr
120 hr
144 hr
168 hr
B
Receptors per Cell
1e1
1e2
1e3
1e4
1e5
1e6
1e7
V Vcell Vnuc Vendo Vfluid Vcelltot Vendotot Vnuctot Ncell q DAR konAg koffAg ki ke frec kiAg keAg frecAg kiADC keADC frecADC keff kinf kinN koutN konDNA koffDNA kg kkill
NaN

Sensitivity analysis changes with time after treatment and receptor expression levels. To examine the sensitivity of the system to the parameters (A) at different times and (B) at varying receptor expression levels, each parameter was varied by 10% and the model was rerun to determine the effect on formation of the effector complex (warhead bound to DNA) and cell population. Values on the heatmap show normalized change in AUC of the effector complex or cell population (% change in AUC / % change in parameter). While many parameters do not cause significant AUC changes, others such as DAR and warhead trafficking rates do, suggesting key design characteristics such as DAR and lipophilicity can be manipulated to alter ADC performance.

Linker Design

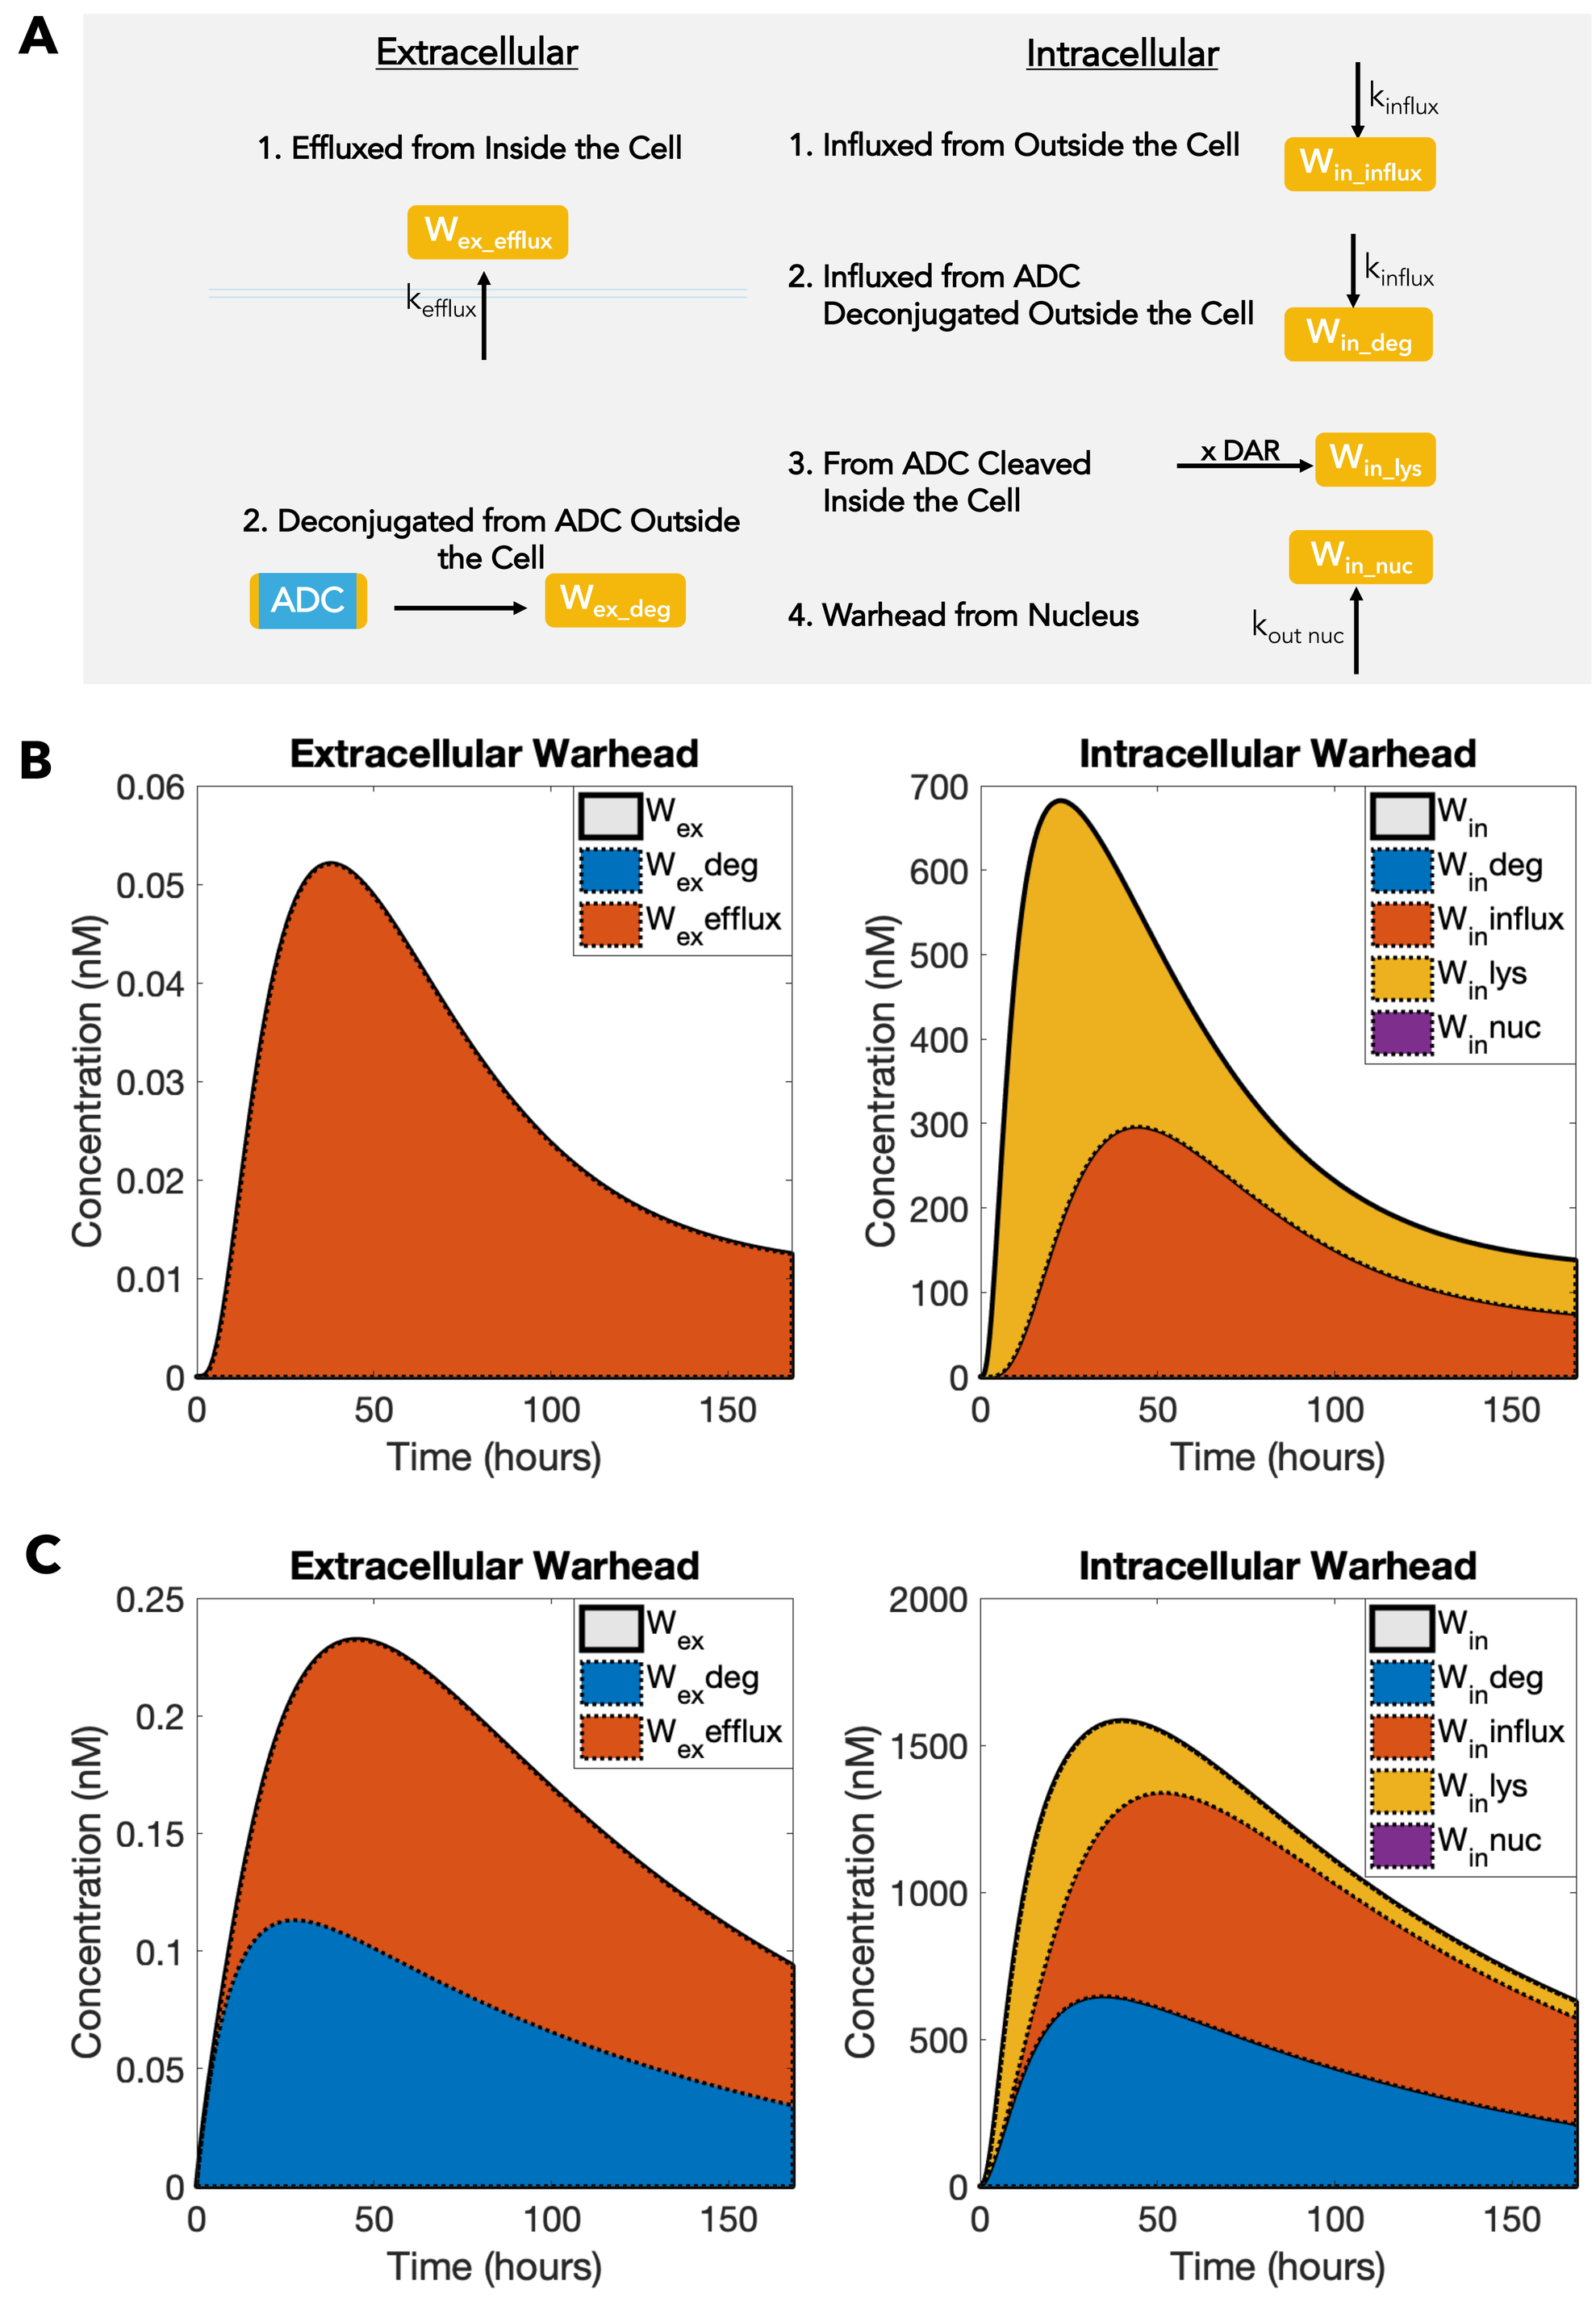
A
Extracellular
1. Effluxed from Inside the Cell
W ex_efflux
k efflux
2. Deconjugated from ADC Outside the Cell
ADC
W ex_deg
Intracellular
1. Influxed from Outside the Cell
k influx
W in_influx
2. Influxed from ADC Deconjugated Outside the Cell
W in_deg
3. From ADC Cleaved Inside the Cell
x DAR
W in_lys
W in_nuc
4. Warhead from Nucleus
k out nuc
B
C
Extracellular Warhead
Intracellular Warhead
Concentration (nM)
Time (hours)
W ex
W ex deg
W ex efflux
W in
W in deg
W in influx
W in lys
W in nuc

Computational model can track warhead location and recent history. Tracking populations of extracellular and intracellular warhead can reveal additional insights into warhead movement and mechanisms of action at the in vitro scale. (A) There are several possible sources for both the extracellular and intracellular warhead subpopulations. (B) For ADCs with noncleavable linkers, the external warhead population is entirely derived from efflux, while intracellular warhead comes from both internal ADC cleavage and influx. A substantial amount of intracellular warhead comes from influx from other cells, suggesting that bystander effects still play a significant role in ADCs with noncleavable linkers. (C) In a hypothetical scenario for ADCs with cleavable linkers, extracellular warhead comes from both external deconjugation and efflux, while intracellular warhead comes from internal cleavage, influx, and external deconjugation.

Conclusions

We have developed a computational systems pharmacology model of PBD-ADCs at the in vitro scale. This model includes both cellular and intracellular mechanisms specific to ADCs with PBD warheads and contains parameters from literature or fit to AZ experimental data. We conduct sensitivity analyses to find the most influential parameters within the system and simulate different scenarios in order to better understand the impact of key ADC characteristics, and connect these to ADC efficacy and toxicity in vitro. Using the relevant experimental data, this in vitro model can be expanded to incorporate whole-body pharmacokinetics, to serve as the basis for in vivo and clinical models of PBD-ADCs. The resulting in models will describe whole-body pharmacology and can be further developed to create a human clinical model, which can be used to run virtual clinical trials.

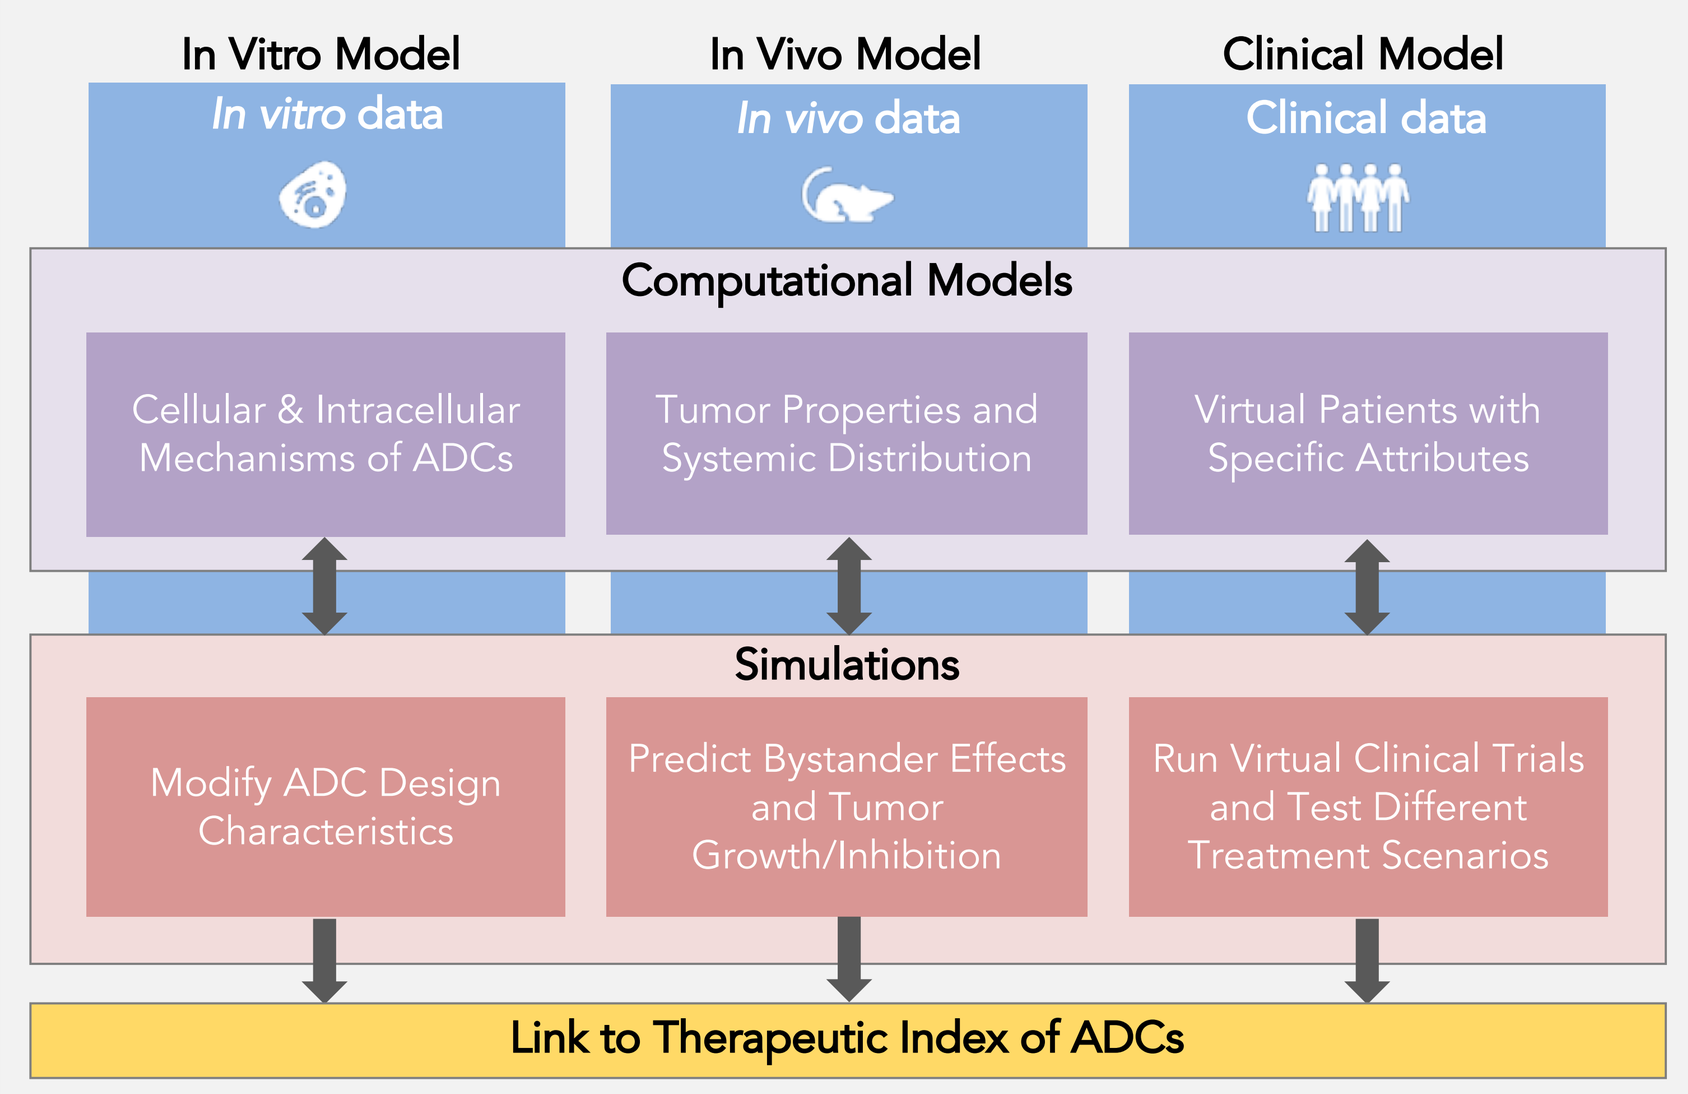
In Vitro Model
In vitro data
In Vivo Model
In vivo data
Clinical Model
Clinical data
Computational Models
Cellular & Intracellular Mechanisms of ADCs
Tumor Properties and Systemic Distribution
Virtual Patients with Specific Attributes
Simulations
Modify ADC Design Characteristics
Predict Bystander Effects and Tumor Growth/Inhibition
Run Virtual Clinical Trials and Test Different Treatment Scenarios
Link to Therapeutic Index of ADCs

Multiscale Model Overview

References

1. Coats, S., et al., *Antibody-Drug Conjugates: Future Directions in Clinical and Translational Strategies to Improve the Therapeutic Index.* Clinical Cancer Research, 2019. **25**(18): p. 5441-5448.
2. Xie, L., E.J. Draizen, and P.E. Bourne, *Harnessing big data for systems pharmacology.* Annual review of pharmacology and toxicology, 2017. **57**: p. 245-262.

Acknowledgments Johns-Hopkins AstraZeneca Scholars Program